80041

ATH-1105, a Small-Molecule Positive Modulator of the HGF/MET System, Is Neuroprotective and Attenuates TDP-43 Protein Pathology in ALS and Frontotemporal Dementia–Relevant Preclinical Models

Jewel L. Johnston, Andrée-Anne Berthiaume, Sherif M. Reda, Kayla N. Kleist, Sharay E. Setti, Wei Wu, Kevin J. Church

Athira Pharma, Inc., Bothell, WA, USA

CONCLUSIONS

Treatment with ATH-1105

- Enhanced neurite outgrowth and synaptogenesis in primary rat hippocampal neurons
- Protected against neurotoxic injury in primary rat cortical neurons
- Mitigated LPS-stimulated cytokine release in THP-1 macrophages
- Attenuated LPS-induced cognitive impairment in vivo
- Reduced markers of inflammation, neurodegeneration, and TDP-43 pathology in a mouse model of ALS and FTD

KEY TAKEAWAY

In preclinical models, ATH-1105 is protective against several pathological mechanisms common to ALS and FTD, supporting its therapeutic potential and continued development for these indications

©Athira Pharma, Inc. All Rights Reserved.

Copies of this poster and supplemental materials, which can be obtained by scanning the QR code, are for personal use only and may not be reproduced without permission from the authors.

Acknowledgments

This study was sponsored by Athira Pharma, Inc. Medical writing support was provided by ApotheCom and funded by Athira Pharma, Inc.

Disclosures

All authors are employees and stock and/or stock options holders of Athira Pharma, Inc.

Disclaimer

ATH-1105 is an investigational therapy that has not received FDA approval and has not been demonstrated to be safe or effective for any use.

Presented at AAIC 2023; July 16-20, 2023; Amsterdam, Netherlands, and virtual

INTRODUCTION

- ALS and FTD are neurodegenerative disorders[1,2] with a high prevalence of TDP-43 protein pathology in affected neurons; approximately 97% and 50% of people with ALS and FTD, respectively, have this protein pathology[3,4]
- Positive modulation of the HGF/MET system has been shown to protect against several drivers of neurodegeneration in preclinical models,[5] and this approach may represent a potential therapeutic strategy for ALS and FTD
- We assessed the effects of ATH-1105, a small-molecule positive modulator of the HGF/MET system, on components of ALS and FTD, including neurodegeneration, inflammation, cognitive impairment, and TDP-43 protein pathology

Figure 1. Positive modulation of the HGF/MET system promotes neurotrophic, neuroprotective, and anti-inflammatory effects

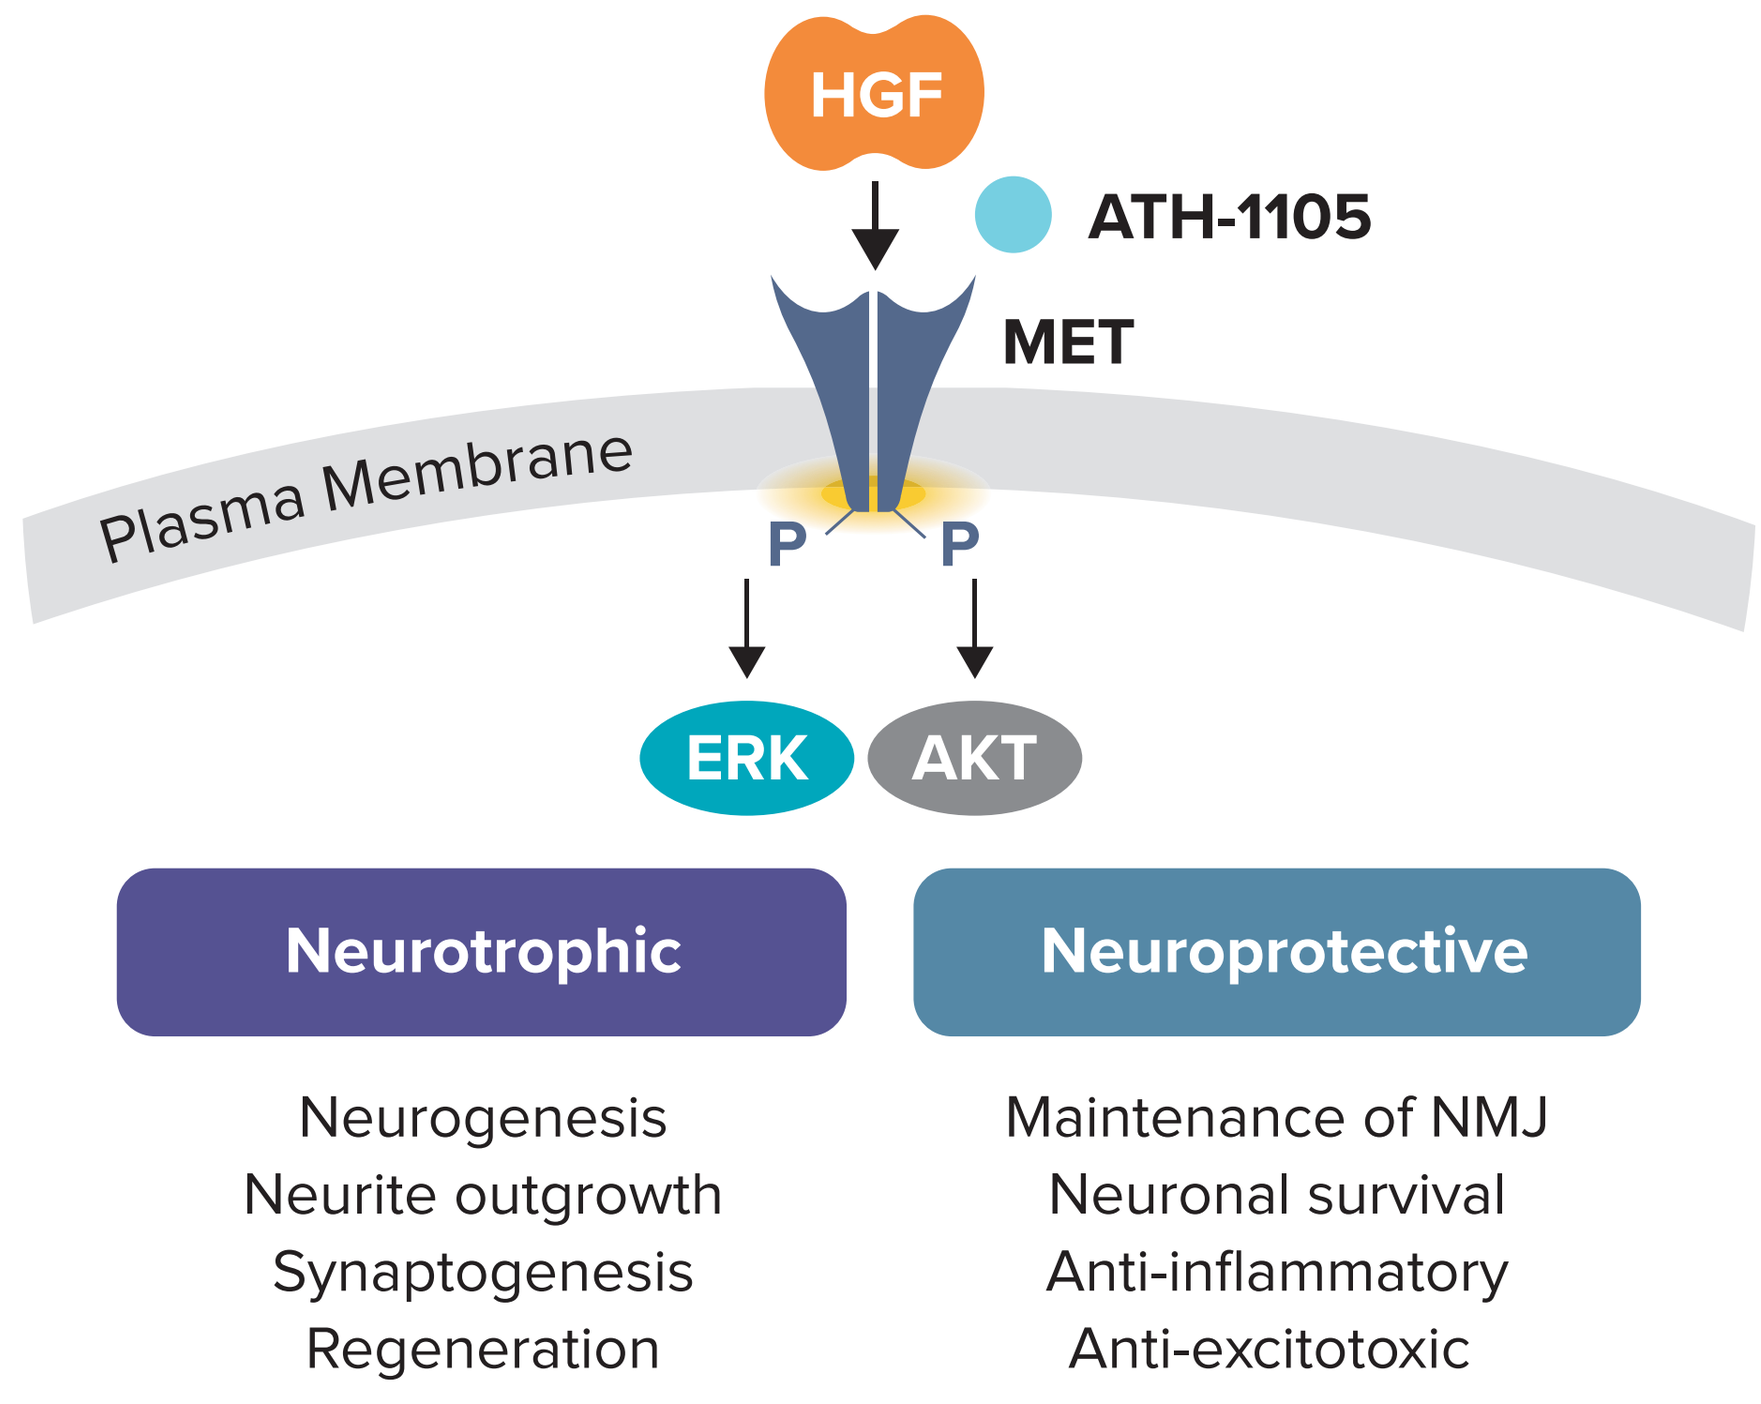
HGF
ATH-1105
MET
Plasma Membrane
P
P
ERK
AKT
Neurotrophic
Neuroprotective
Neurogenesis
Neurite outgrowth
Synaptogenesis
Regeneration
Maintenance of NMJ
Neuronal survival
Anti-inflammatory
Anti-excitotoxic

OBJECTIVE

To assess the potential neurotrophic, neuroprotective, and anti-inflammatory effects of ATH-1105 in preclinical models relevant to ALS and FTD

METHODS

Neurotrophic and neuroprotective effects in primary cell culture

- Primary rat hippocampal neurons were treated with ATH-1105 every other day for 3 days or 9 days. Neurite outgrowth was assessed on Day 3 and synaptogenesis was assessed on Day 9
- Cell viability of primary rat cortical neurons was measured after treatment with ATH-1105 for 15 minutes, followed by neurotoxic injury with glutamate, LPS, H_2O_2, or MPP^+ for 24 hours

In vitro LPS-induced proinflammatory cytokine release

- THP-1–differentiated macrophages were pretreated with ATH-1105 for 20 minutes, then challenged with LPS 50 ng/mL for 24 hours. All cultures included 0.5 ng/mL HGF. Proinflammatory cytokine levels were evaluated by HTRF

In vivo LPS-induced cognitive impairment assay

- Adult mice received a single IP injection of LPS 0.25 mg/kg, followed by ATH-1105 PO QD for 14 days; cognitive performance was then assessed using the T-maze spontaneous alternation test

In vivo Prp-TDP43^{A315T} model of ALS and FTD

- Prp-TDP43^{A315T} (JAX strain #010700) mice were given ATH-1105 10 mg/kg or vehicle PO QD from 1 to 3 months of age (2 months of treatment). Plasma levels of cytokines and NfL were measured by ELISA. pTDP-43 inclusions in the sciatic nerve were evaluated by IHC using anti–pTDP-43 (Ser409/410) antibody. Intensity of pTDP-43 labeling was quantified

RESULTS

Figure 2. ATH-1105 enhances neurite outgrowth and synaptogenesis in primary rat hippocampal neurons

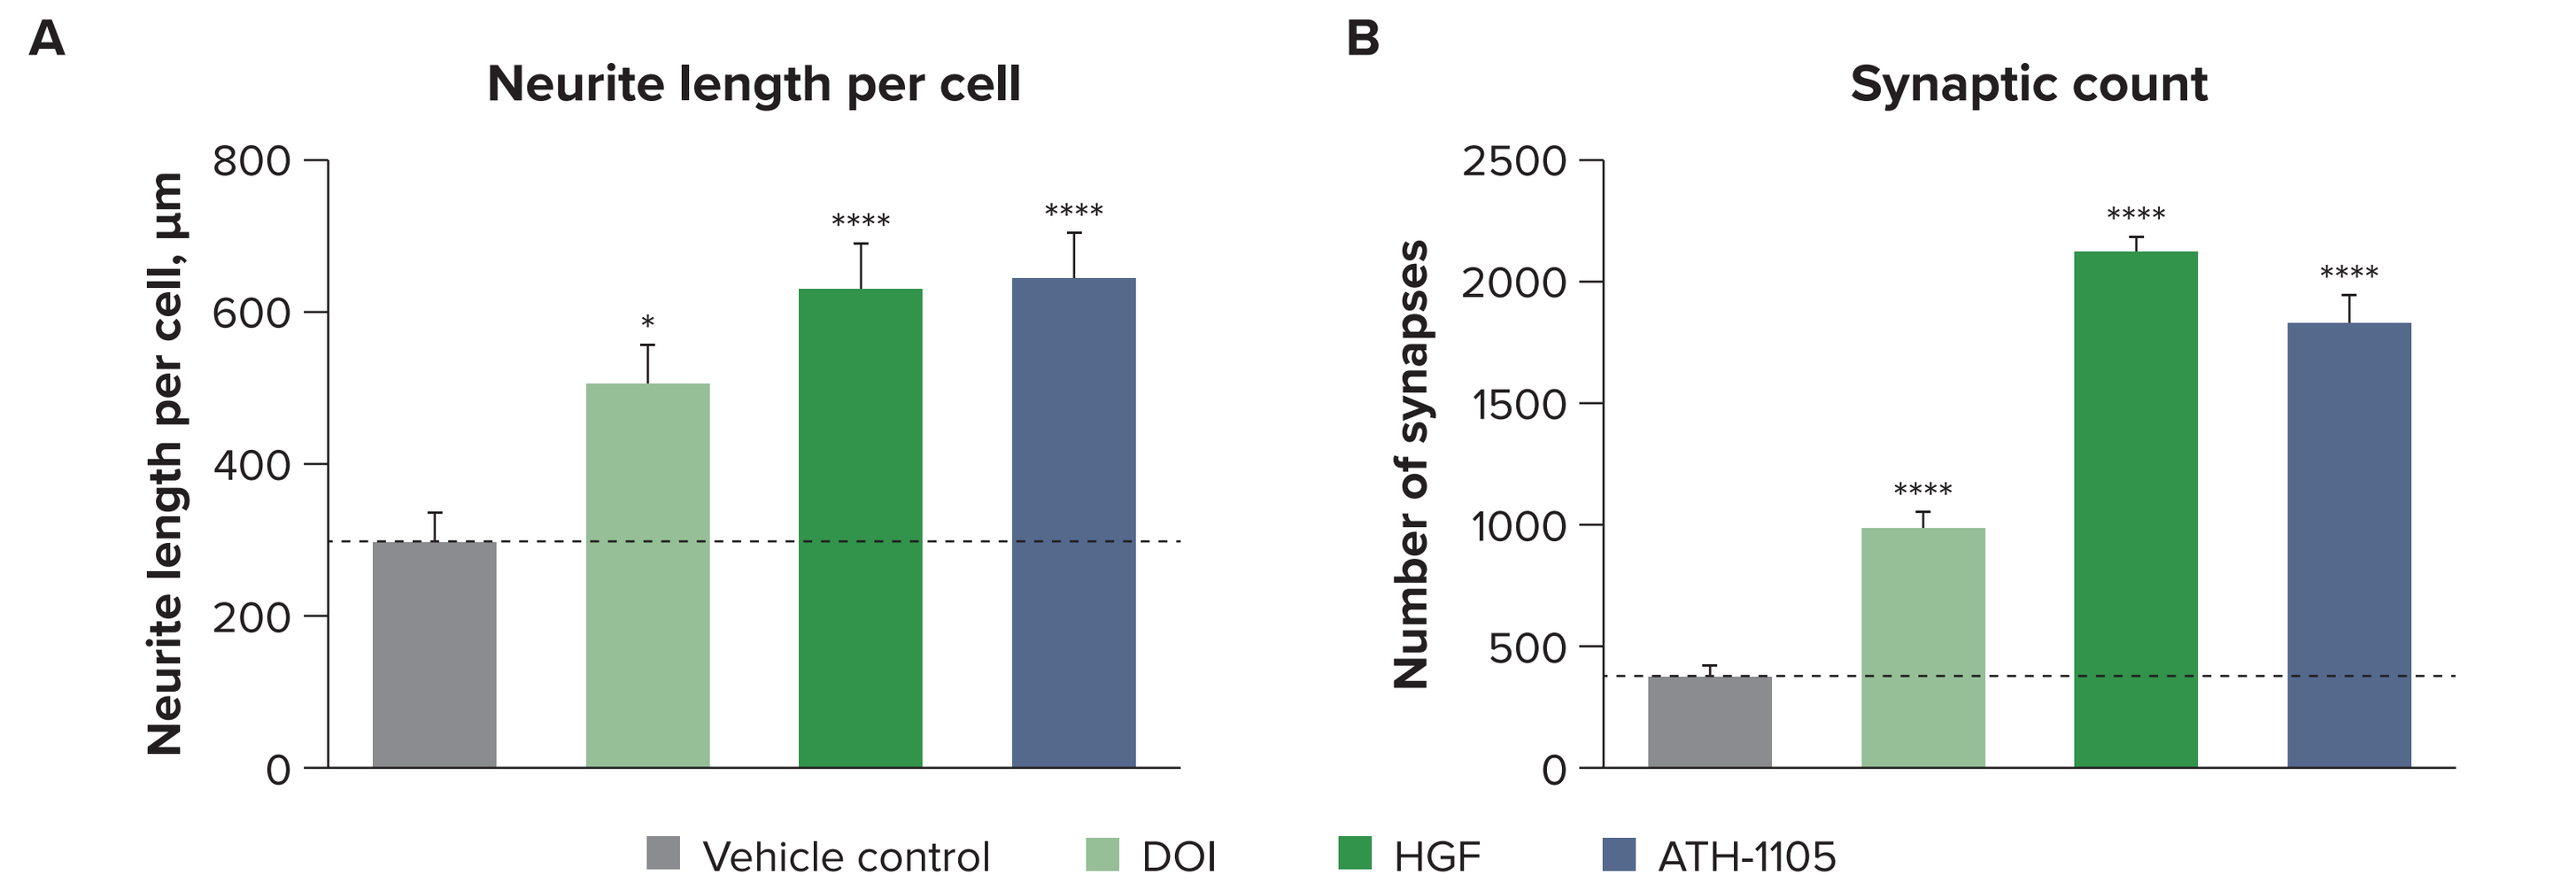
A
Neurite length per cell
Neurite length per cell, µm
0
200
400
600
800
*

B
Synaptic count
Number of synapses
0
500
1000
1500
2000
2500

Vehicle control
DOI
HGF
ATH-1105

Graphical representation of (A) neurite length per cell and (B) synaptic count with DOI, HGF 5 ng/mL, or ATH-1105 1 nM treatment.

Data presented as mean + SEM; n = 10 images analyzed for each treatment condition, from n = 3 wells per group. One-way ANOVA with Dunnett's test vs vehicle.

*$p < 0.05$, ****$p < 0.0001$.

Figure 3. ATH-1105 protects primary rat cortical neurons from several neurotoxic injuries

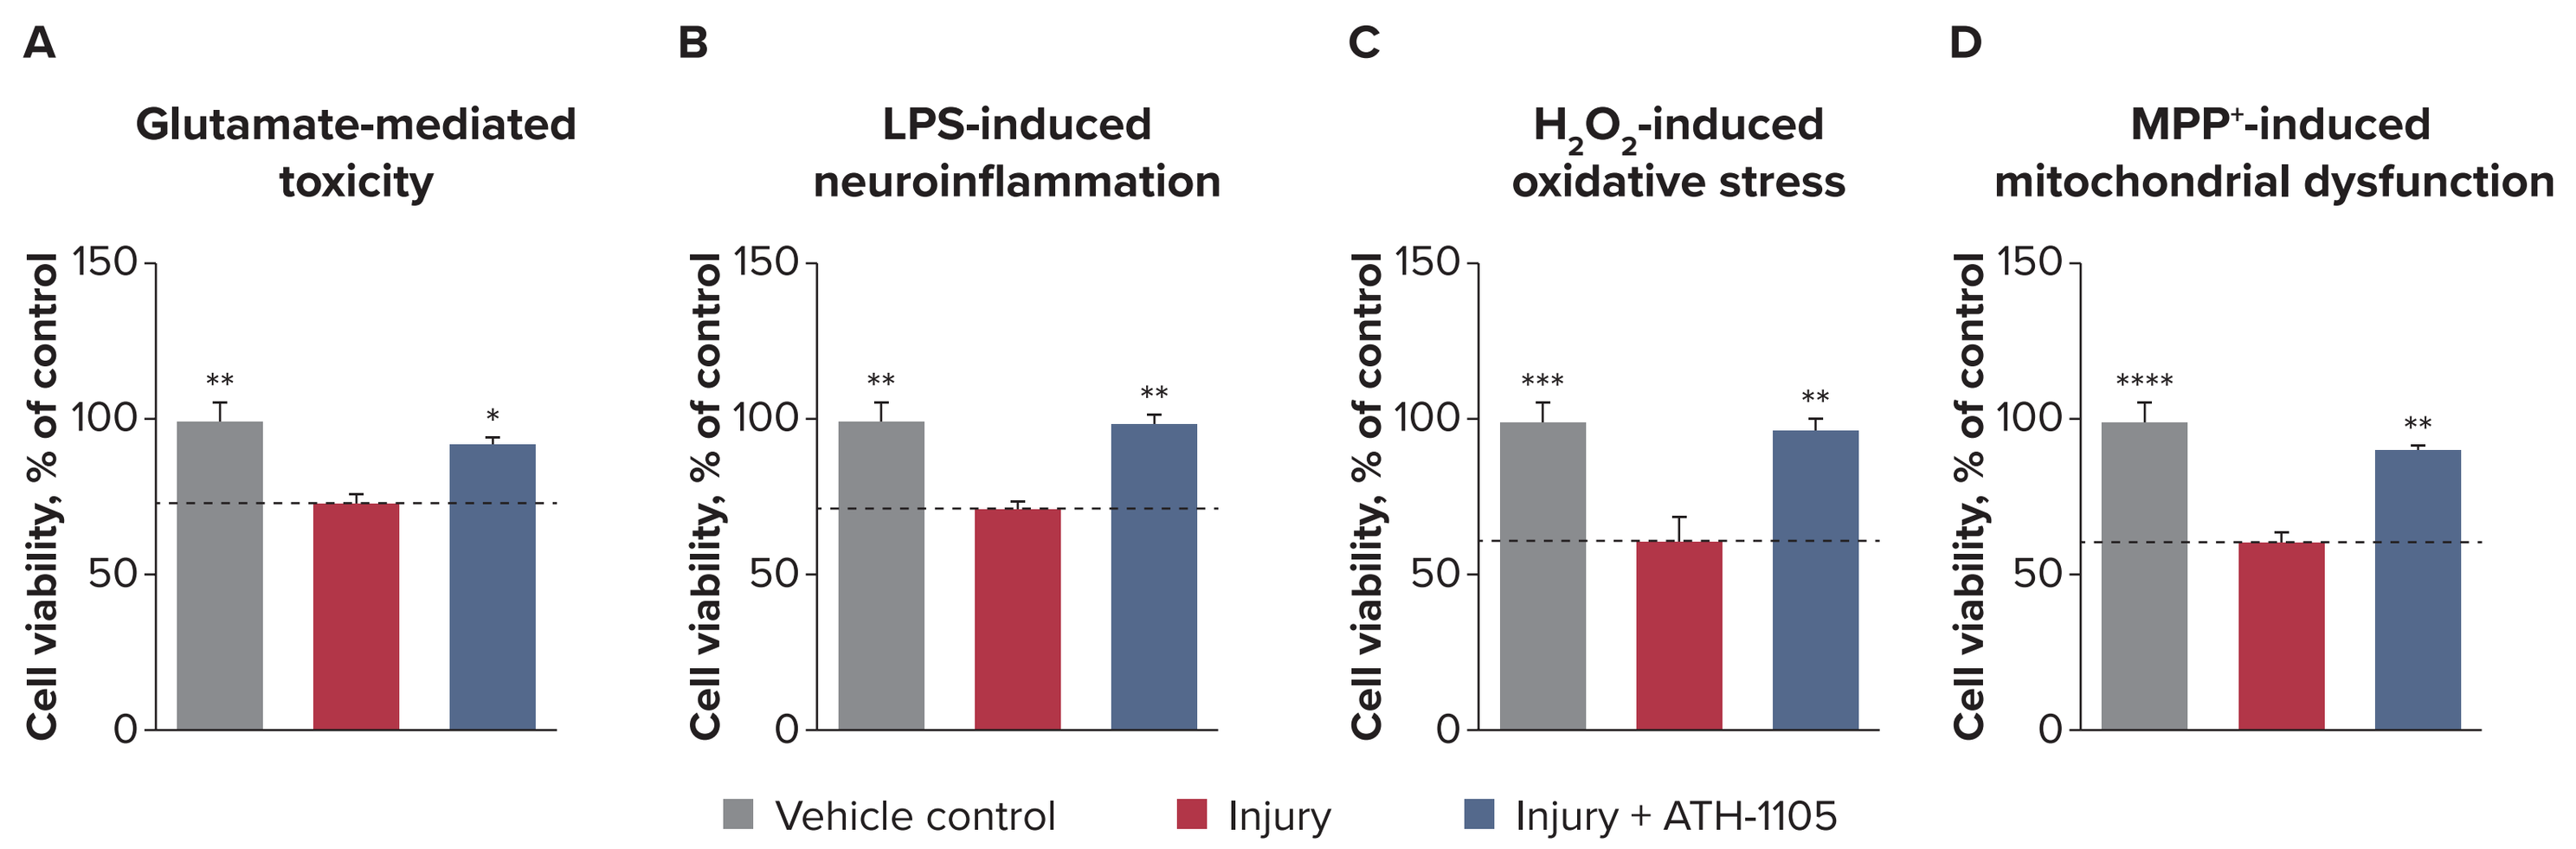
A
Glutamate-mediated toxicity
B
LPS-induced neuroinflammation
C
H_2O_2-induced oxidative stress
D
MPP^+-induced mitochondrial dysfunction
Cell viability, % of control
0
50
100
150
**
*

Vehicle control
Injury
Injury + ATH-1105

Percentage of primary rat cortical neurons surviving in culture after 24 h exposure to (A) glutamate (25 µM), (B) LPS (1 µM), (C) H_2O_2 (1 µM), and (D) MPP^+ (500 µM). ATH-1105 dose was 1 nM.

Data presented as mean + SEM; vehicle control group, n = 6; injury groups, n = 4 each. One-way ANOVA with Dunnett's test vs injury.

*$p < 0.05$, **$p < 0.01$, ***$p < 0.001$, ****$p < 0.0001$.

Figure 4. ATH-1105 reduces LPS-induced proinflammatory cytokine release in THP-1 macrophages

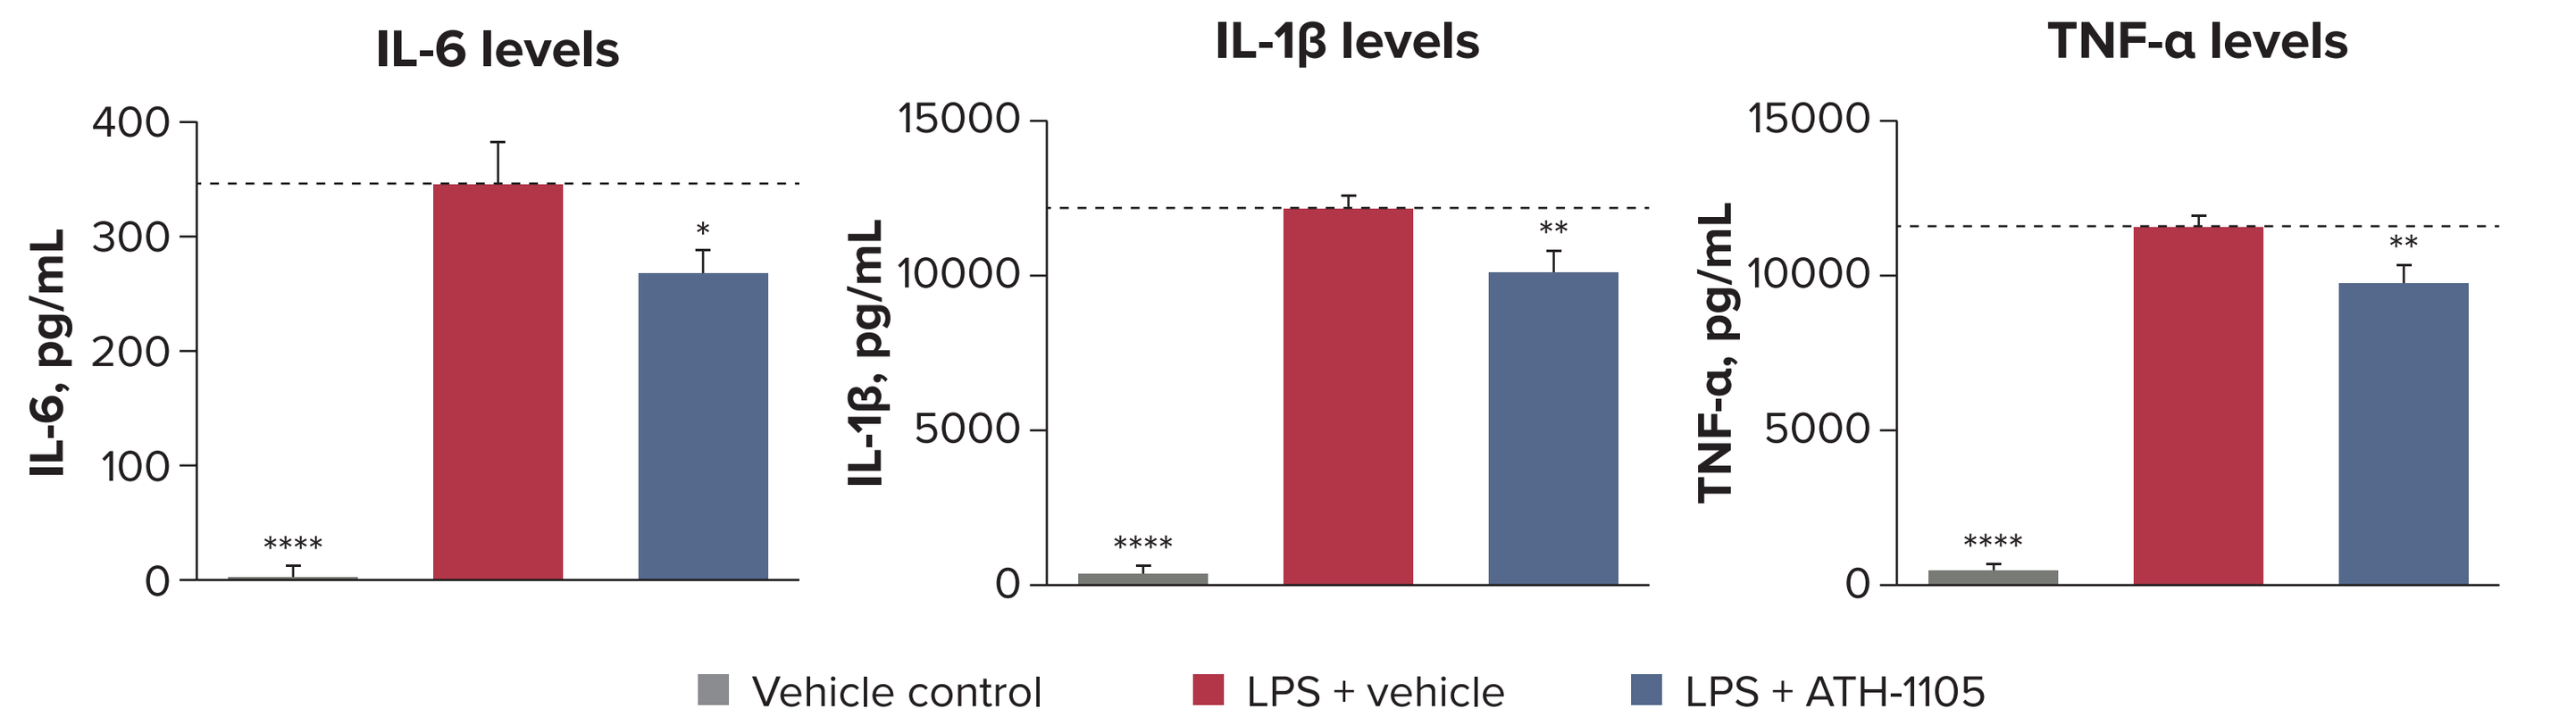
IL-6 levels
IL-6, pg/mL
0
100
200
300
400

*
IL-1β levels
IL-1β, pg/mL
0
5000
10000
15000

**
TNF-α levels
TNF-α, pg/mL

**
Vehicle control
LPS + vehicle
LPS + ATH-1105

Levels of (A) IL-6, (B) IL-1β, and (C) TNF-α release after LPS challenge with vehicle or ATH-1105 1 nM.

Data presented as mean + SEM; n = 6 wells per group. One-way ANOVA with Dunnett's test vs LPS + vehicle.

*$p < 0.05$, **$p < 0.01$, ****$p < 0.0001$.

Figure 5. ATH-1105 attenuates LPS-induced cognitive impairment in mice

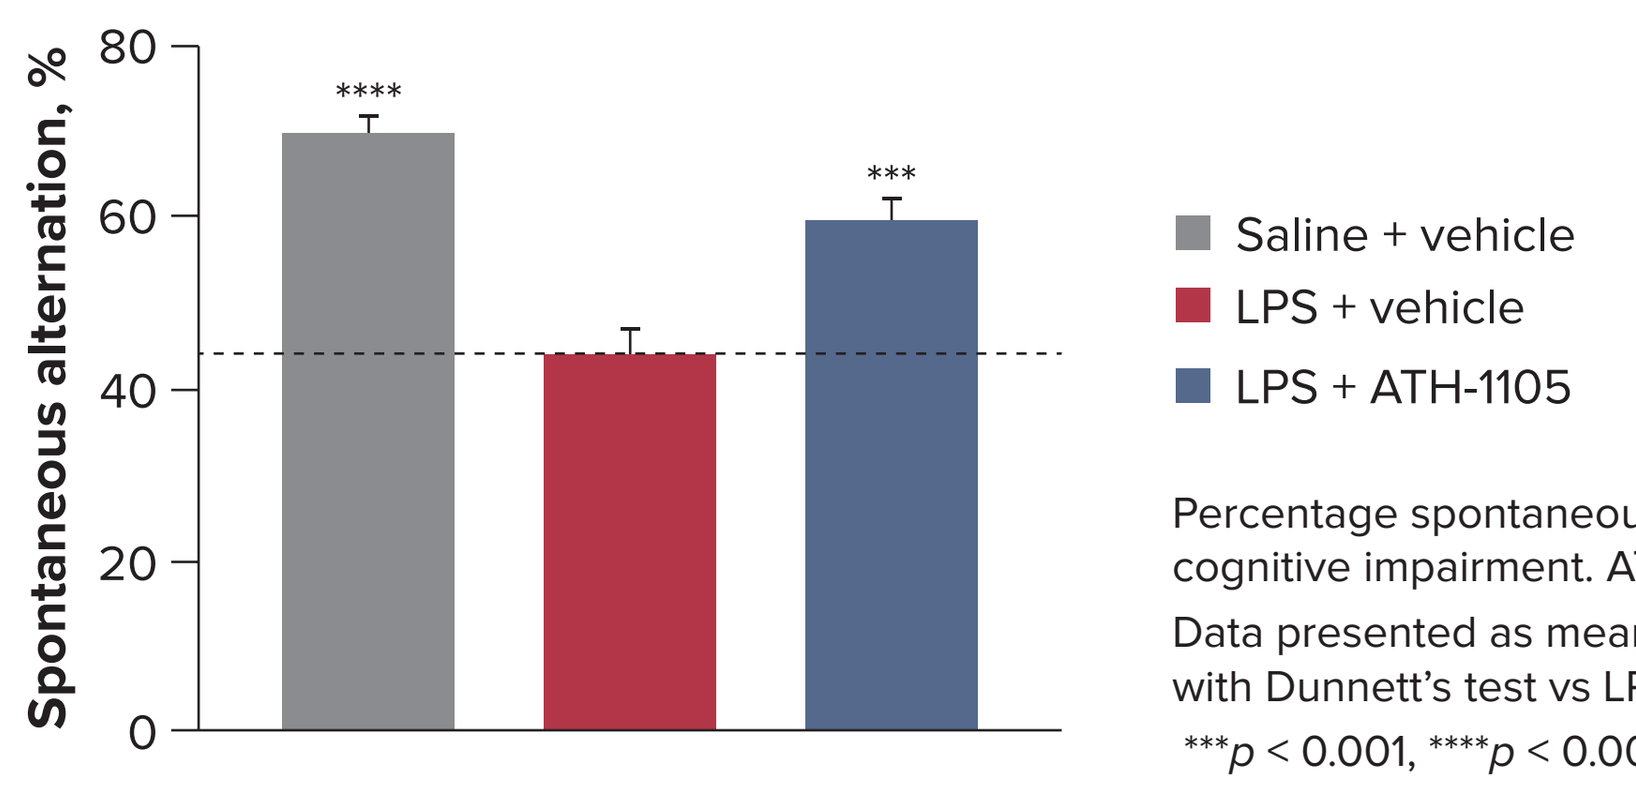
Cognitive performance in T-maze
Spontaneous alternation, %
0
20
40
60
80

Saline + vehicle
LPS + vehicle
LPS + ATH-1105

Percentage spontaneous alternations in the T-maze in an LPS-induced mouse model of cognitive impairment. ATH-1105 dose was 20 mg/kg.

Data presented as mean + SEM; n = 10 mice per group. One-way ANOVA with Dunnett's test vs LPS + vehicle.

$p < 0.001$, *$p < 0.0001$.

Figure 6. ATH-1105 reduces markers of inflammation and neurodegeneration in the Prp-TDP43^{A315T} mouse model of ALS and FTD

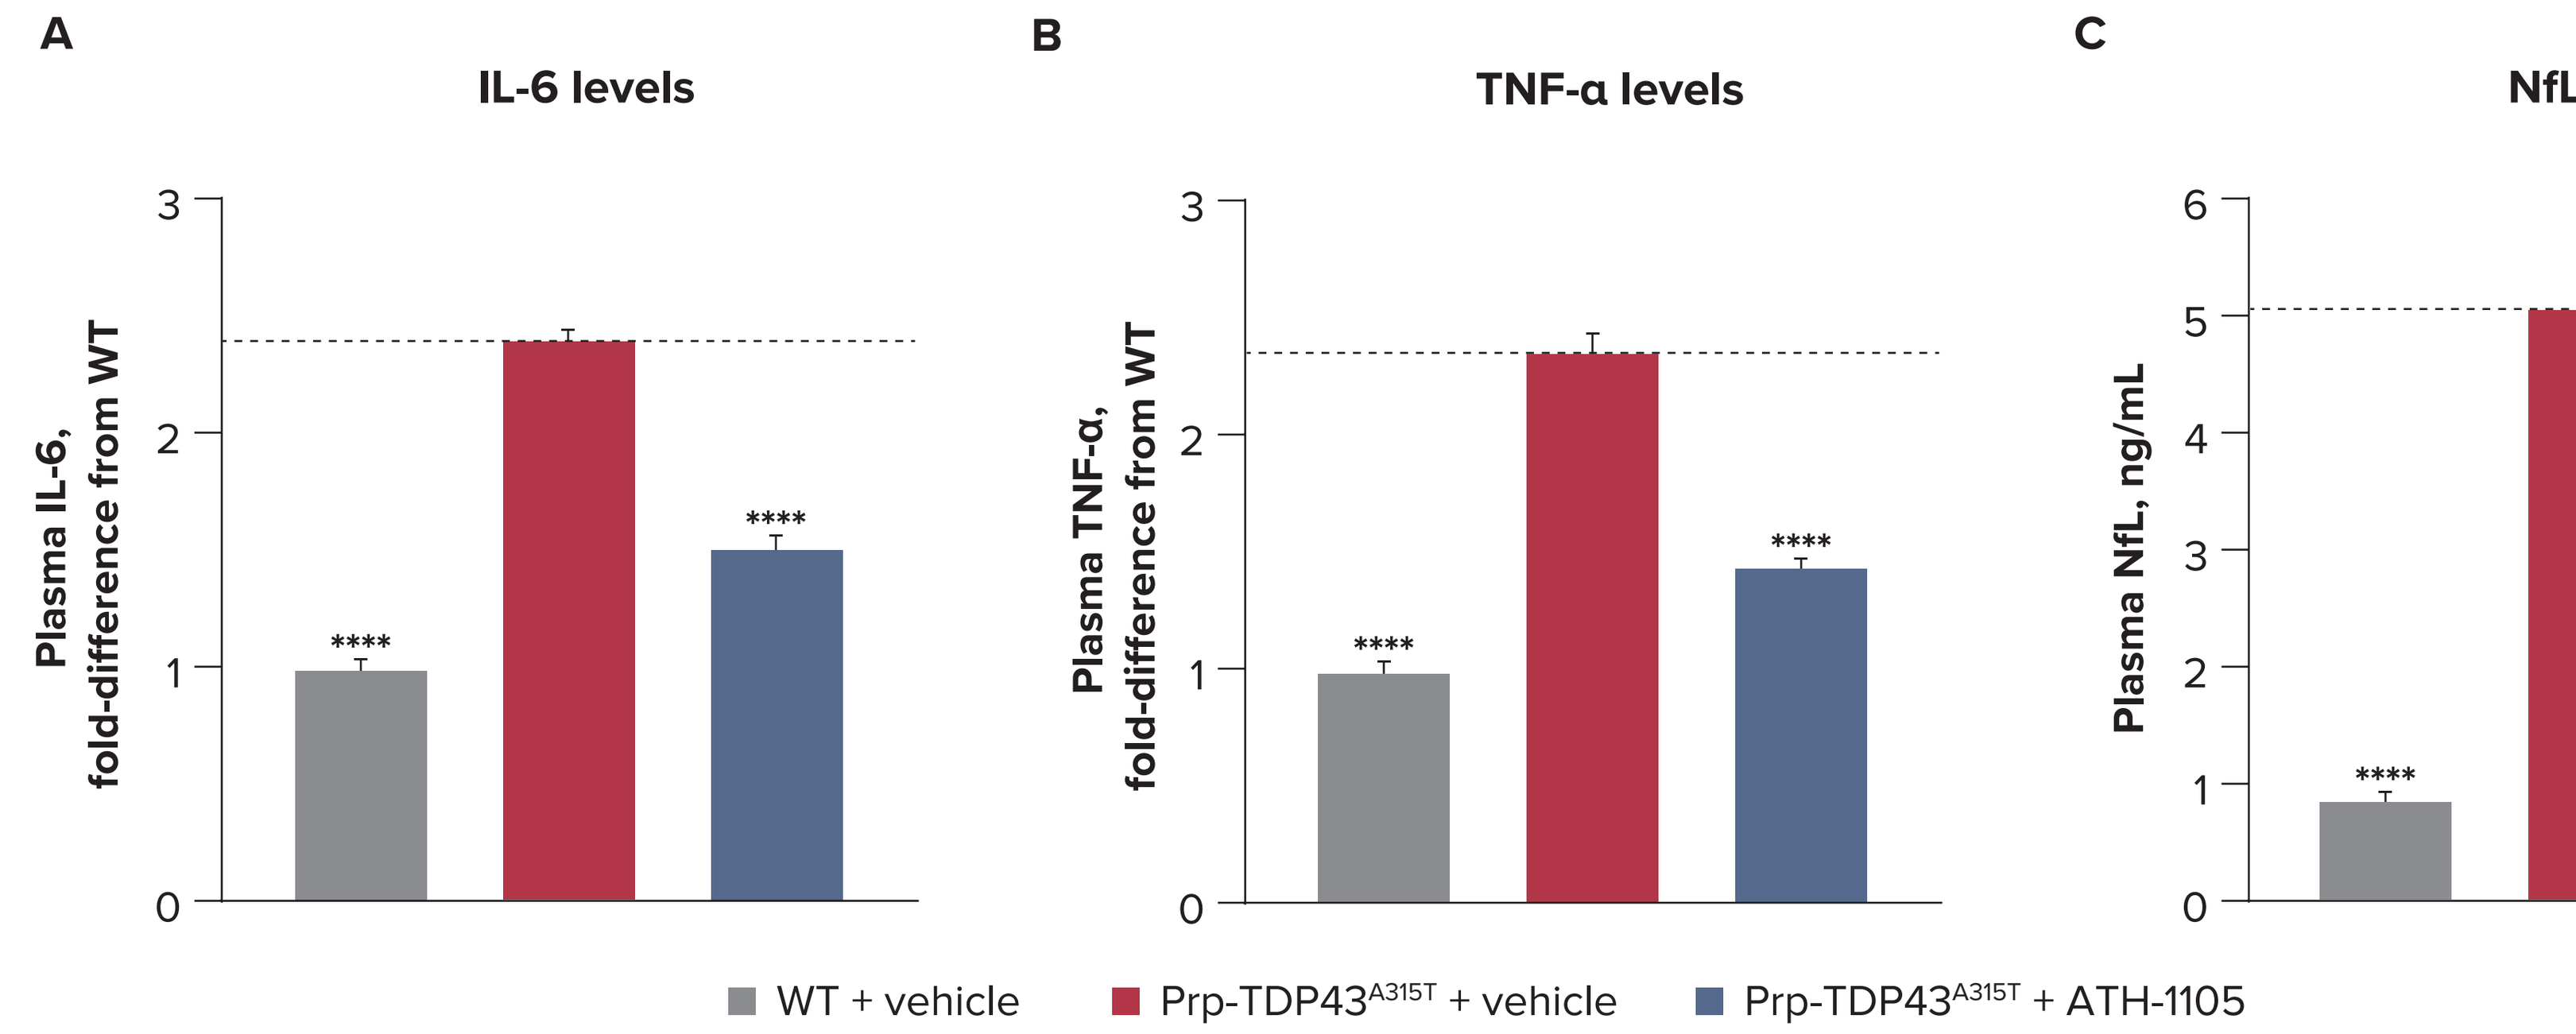
A
IL-6 levels
Plasma IL-6, fold-difference from WT
0
1
2
3

B
TNF-α levels
Plasma TNF-α, fold-difference from WT

C
NfL levels
Plasma NfL, ng/mL
0
1
2
3
4
5
6

WT + vehicle
Prp-TDP43^{A315T} + vehicle
Prp-TDP43^{A315T} + ATH-1105

Graphical representation of plasma (A) IL-6 and (B) TNF-α in fold-difference from WT + vehicle, and (C) NfL concentration after 2 months of treatment. ATH-1105 dose was 10 mg/kg.

Data presented as mean + SEM; n = 10 mice per group. One-way ANOVA with Dunnett's test vs Prp-TDP43^{A315T} + vehicle.

****$p < 0.0001$.

Figure 7. ATH-1105 reduces pTDP-43 levels in the sciatic nerve of Prp-TDP43^{A315T} mice after 2 months of treatment

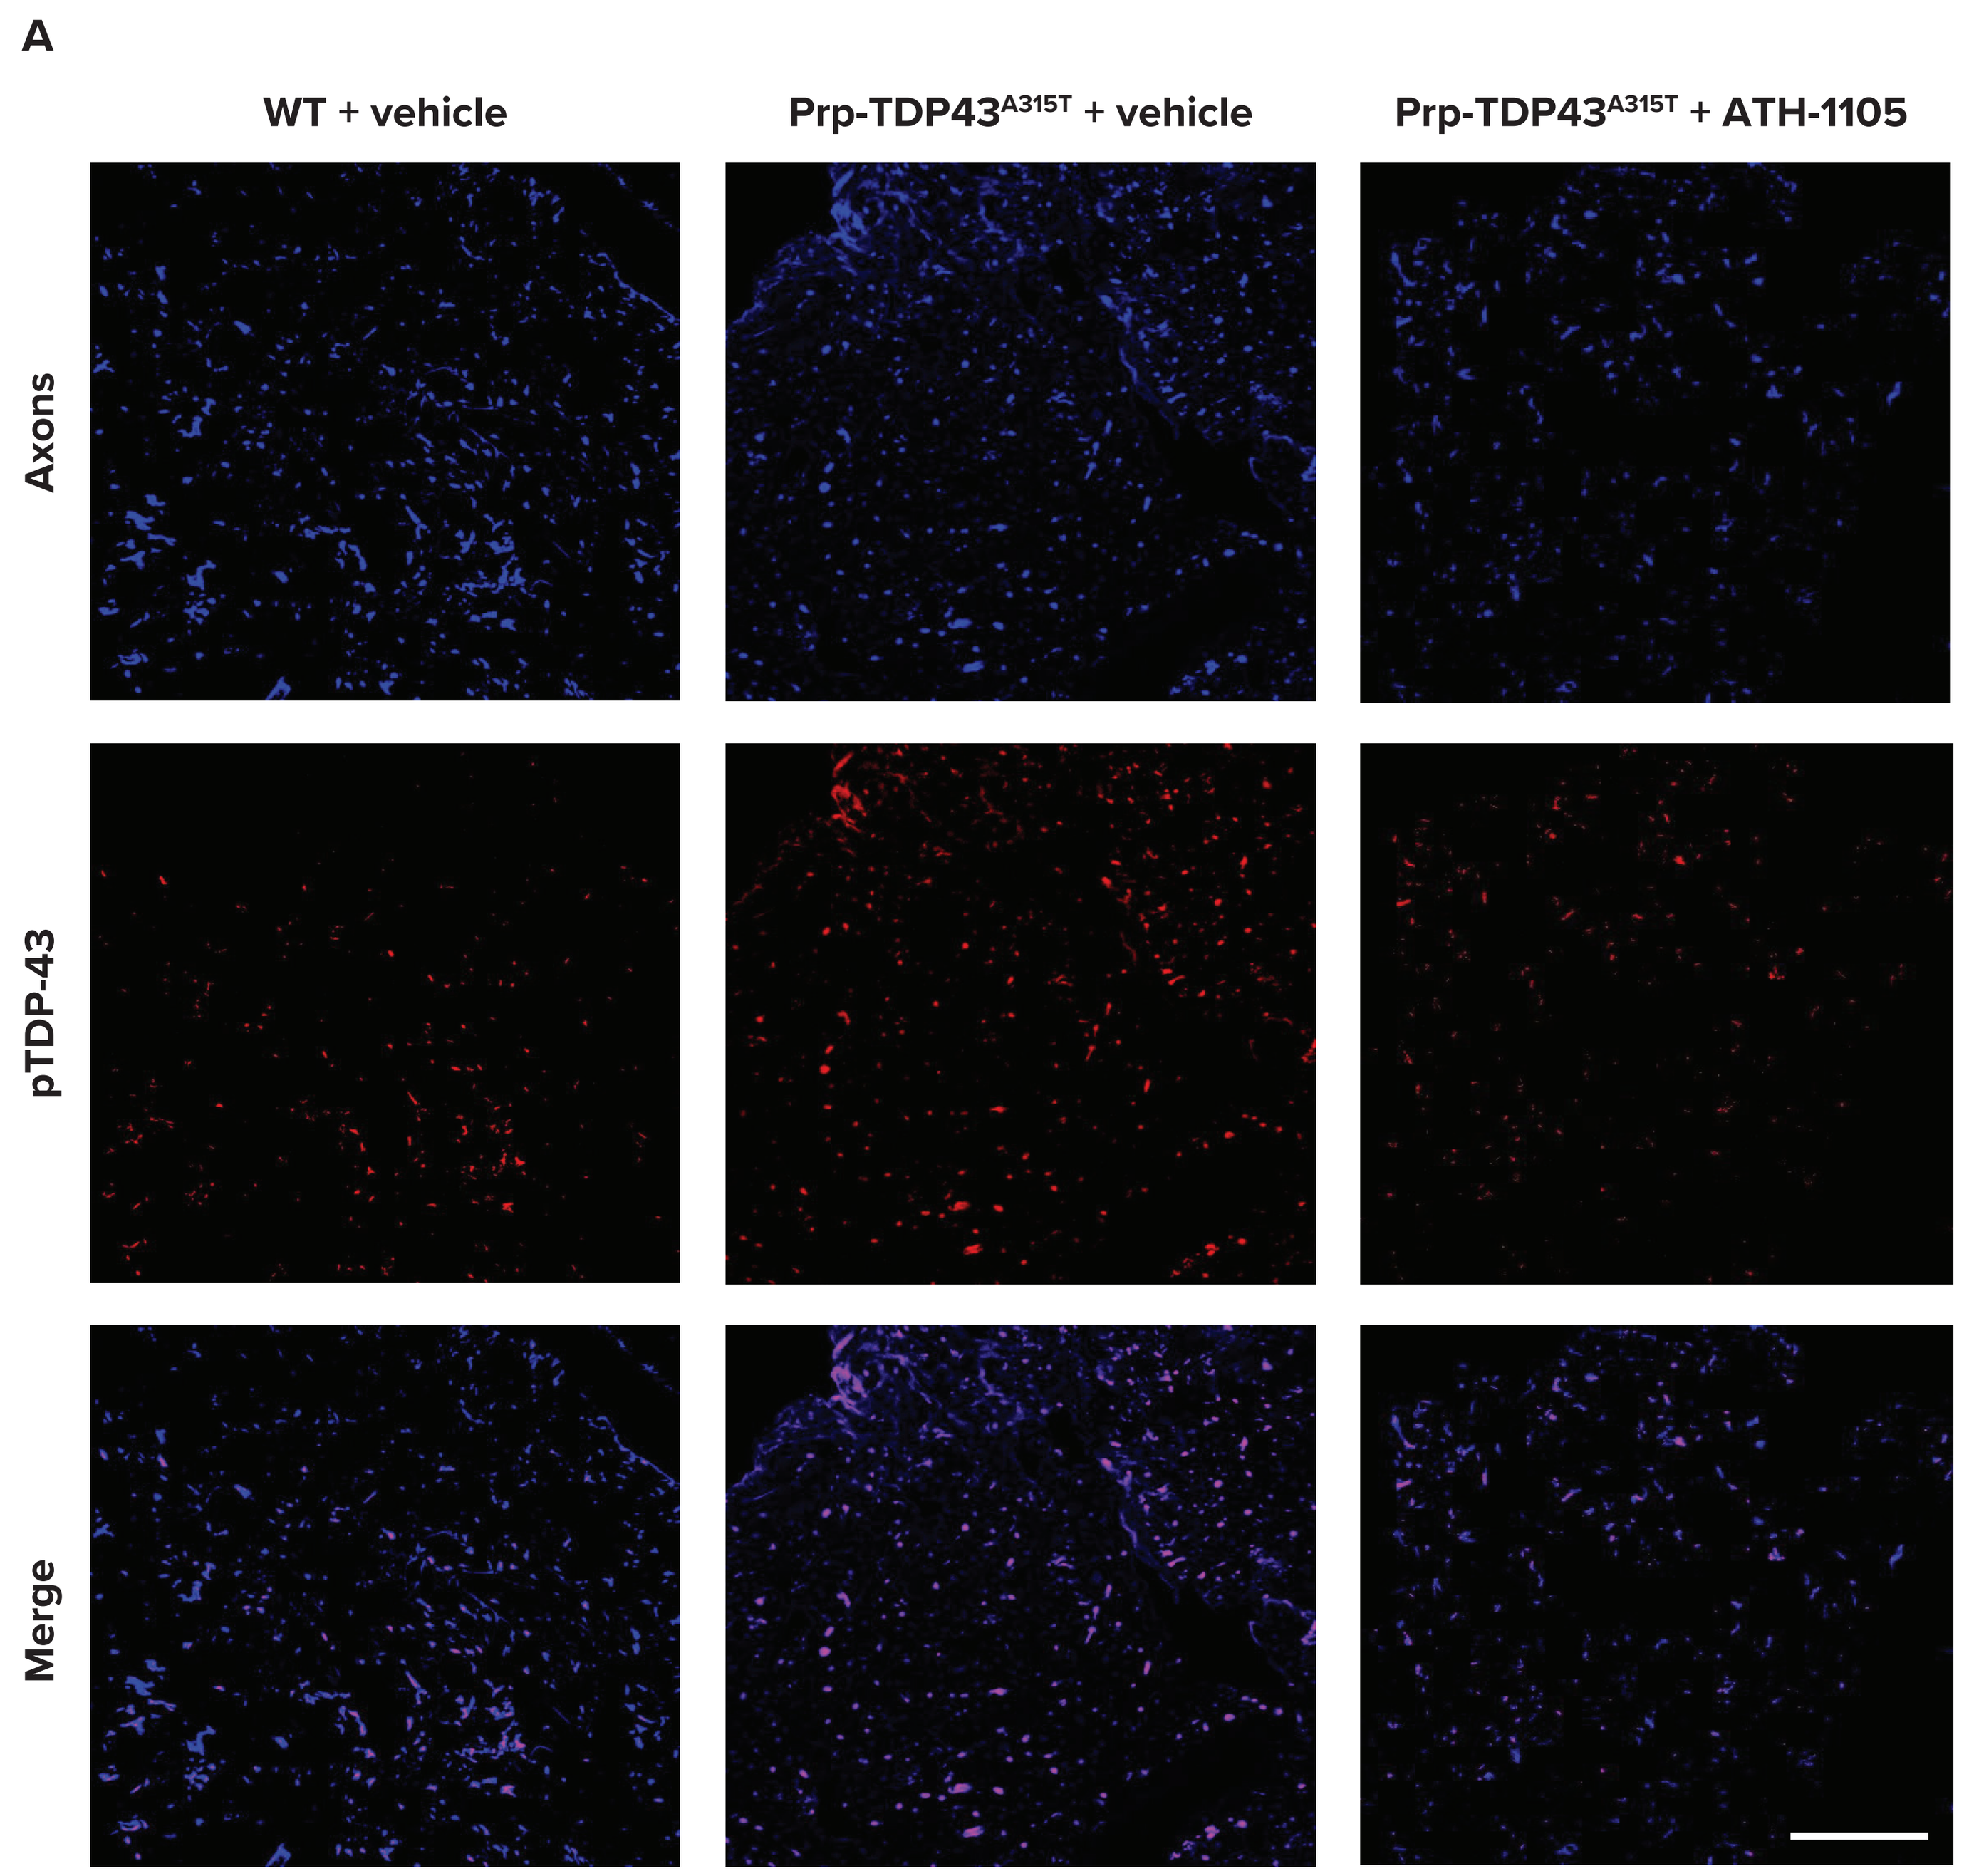
A
WT + vehicle
Prp-TDP43^{A315T} + vehicle
Prp-TDP43^{A315T} + ATH-1105
Axons
pTDP-43
Merge

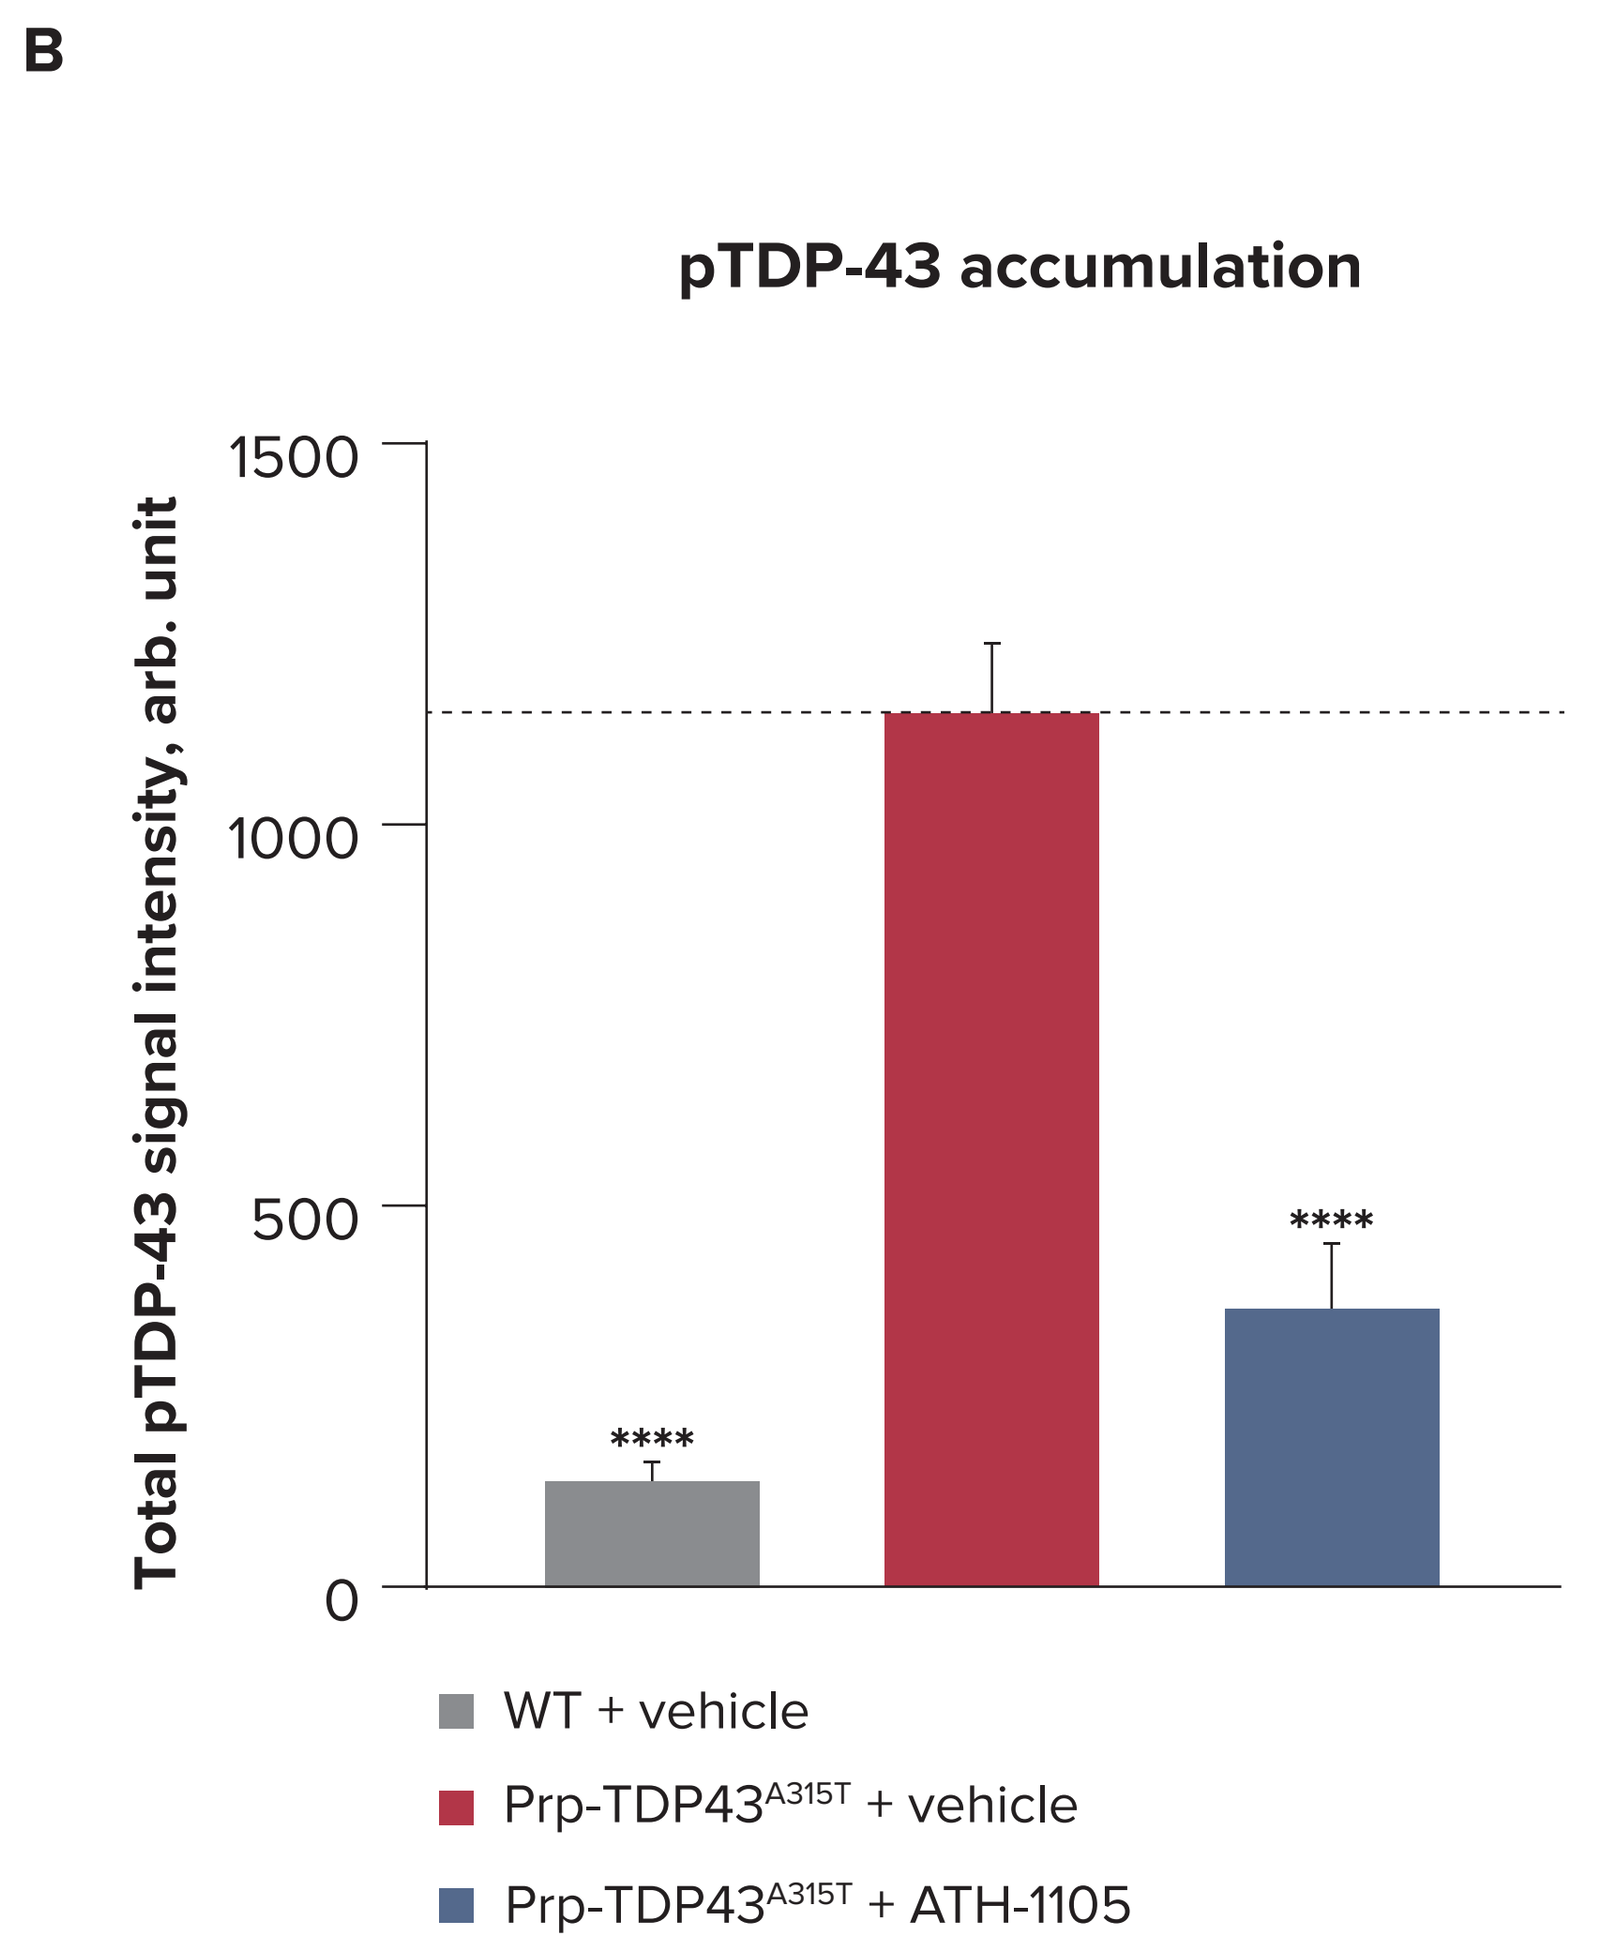
B
pTDP-43 accumulation
Total pTDP-43 signal intensity, arb. unit
0
500
1000
1500

WT + vehicle
Prp-TDP43^{A315T} + vehicle
Prp-TDP43^{A315T} + ATH-1105

(A) Representative images (scale bar = 50 µm) of 10-µm thick cross-sections of sciatic nerves labeled for axons (Tuj1) and pTDP-43. (B) Quantification of ATH-1105 effects on extranuclear pTDP-43 accumulation. ATH-1105 dose was 20 mg/kg.

Data presented as mean + SEM; n = 7 mice per group. One-way ANOVA with Dunnett's test vs Prp-TDP43^{A315T} + vehicle.

****$p < 0.0001$.

Abbreviations **AKT**, protein kinase B; **ALS**, amyotrophic lateral sclerosis; **ANOVA**, analysis of variance; **arb. unit**, arbitrary unit; **DOI**, (±)-2,5-Dimethoxy-4-iodoamphetamine hydrochloride; **ELISA**, enzyme-linked immunosorbent assay; **ERK**, extracellular signal–regulated kinase; **FTD**, frontotemporal dementia; **H_2O_2**, hydrogen peroxide; **HGF**, hepatocyte growth factor; **HTRF**, homogeneous time-resolved fluorescence; **IHC**, immunohistochemistry; **IL-6**, interleukin 6; **IL-1β**, interleukin-1β; **IP**, intraperitoneal; **LPS**, lipopolysaccharide; **MPP^+**, 1-methyl-4-phenylpyridium; **NfL**, neurofilament light chain; **NMJ**, neuromuscular junction; **P**, phosphorylation; **PO**, orally; **PrP**, mouse prion protein; **pTDP-43**, phosphorylated TAR DNA-binding protein 43; **QD**, once daily; **SEM**, standard error of the mean; **TDP-43**, TAR DNA-binding protein 43; **THP-1**, human leukemia monocyte cell line; **TNF-α**, tumor necrosis factor α; **Tuj1**, beta-III tubulin; **WT**, wild type.

References **1.** Bang J et al. *Lancet*. 2015;386(10004):1672-1682. **2.** Tortelli R et al. *Front Neurol*. 2020;11:552295. **3.** Wood A et al. *Int J Mol Sci*. 2021;22(9):4705. **4.** Scotter EL et al. *Neurotherapeutics*. 2015;12(2):352-363. **5.** Johnston JL et al. *Neurotherapeutics*. 2023;20(2):431-451.

80041

ATH-1105, a Small-Molecule Positive Modulator of the HGF/MET System, Is Neuroprotective and Attenuates TDP-43 Protein Pathology in ALS and Frontotemporal Dementia–Relevant Preclinical Models

Jewel L. Johnston, Andrée-Anne Berthiaume, Sherif M. Reda, Kayla N. Kleist, Sharay E. Setti, Wei Wu, Kevin J. Church

Athira Pharma, Inc., Bothell, WA, USA

SUPPLEMENTAL INFORMATION

MET activation assay

- HEK293 cells were incubated with vehicle control, HGF 1 ng/mL, or HGF 1 ng/mL + ATH-1105 100 pM in 6-well plates for 15 minutes
- Cells were lysed, and levels of pMET stimulated by each treatment were measured via ELISA

AKT/ERK activation assay

- HEK293 cells were incubated with vehicle control (containing HGF 2 ng/mL) or ATH-1105 1 μM in 96-well plates for 20 minutes
- Cell lysates were immunolabeled using anti-pAKT or anti-pERK antibodies and quantified using an HTRF reader

Supplemental Figure S1. ATH-1105 enhances MET, AKT, and ERK activation in vitro

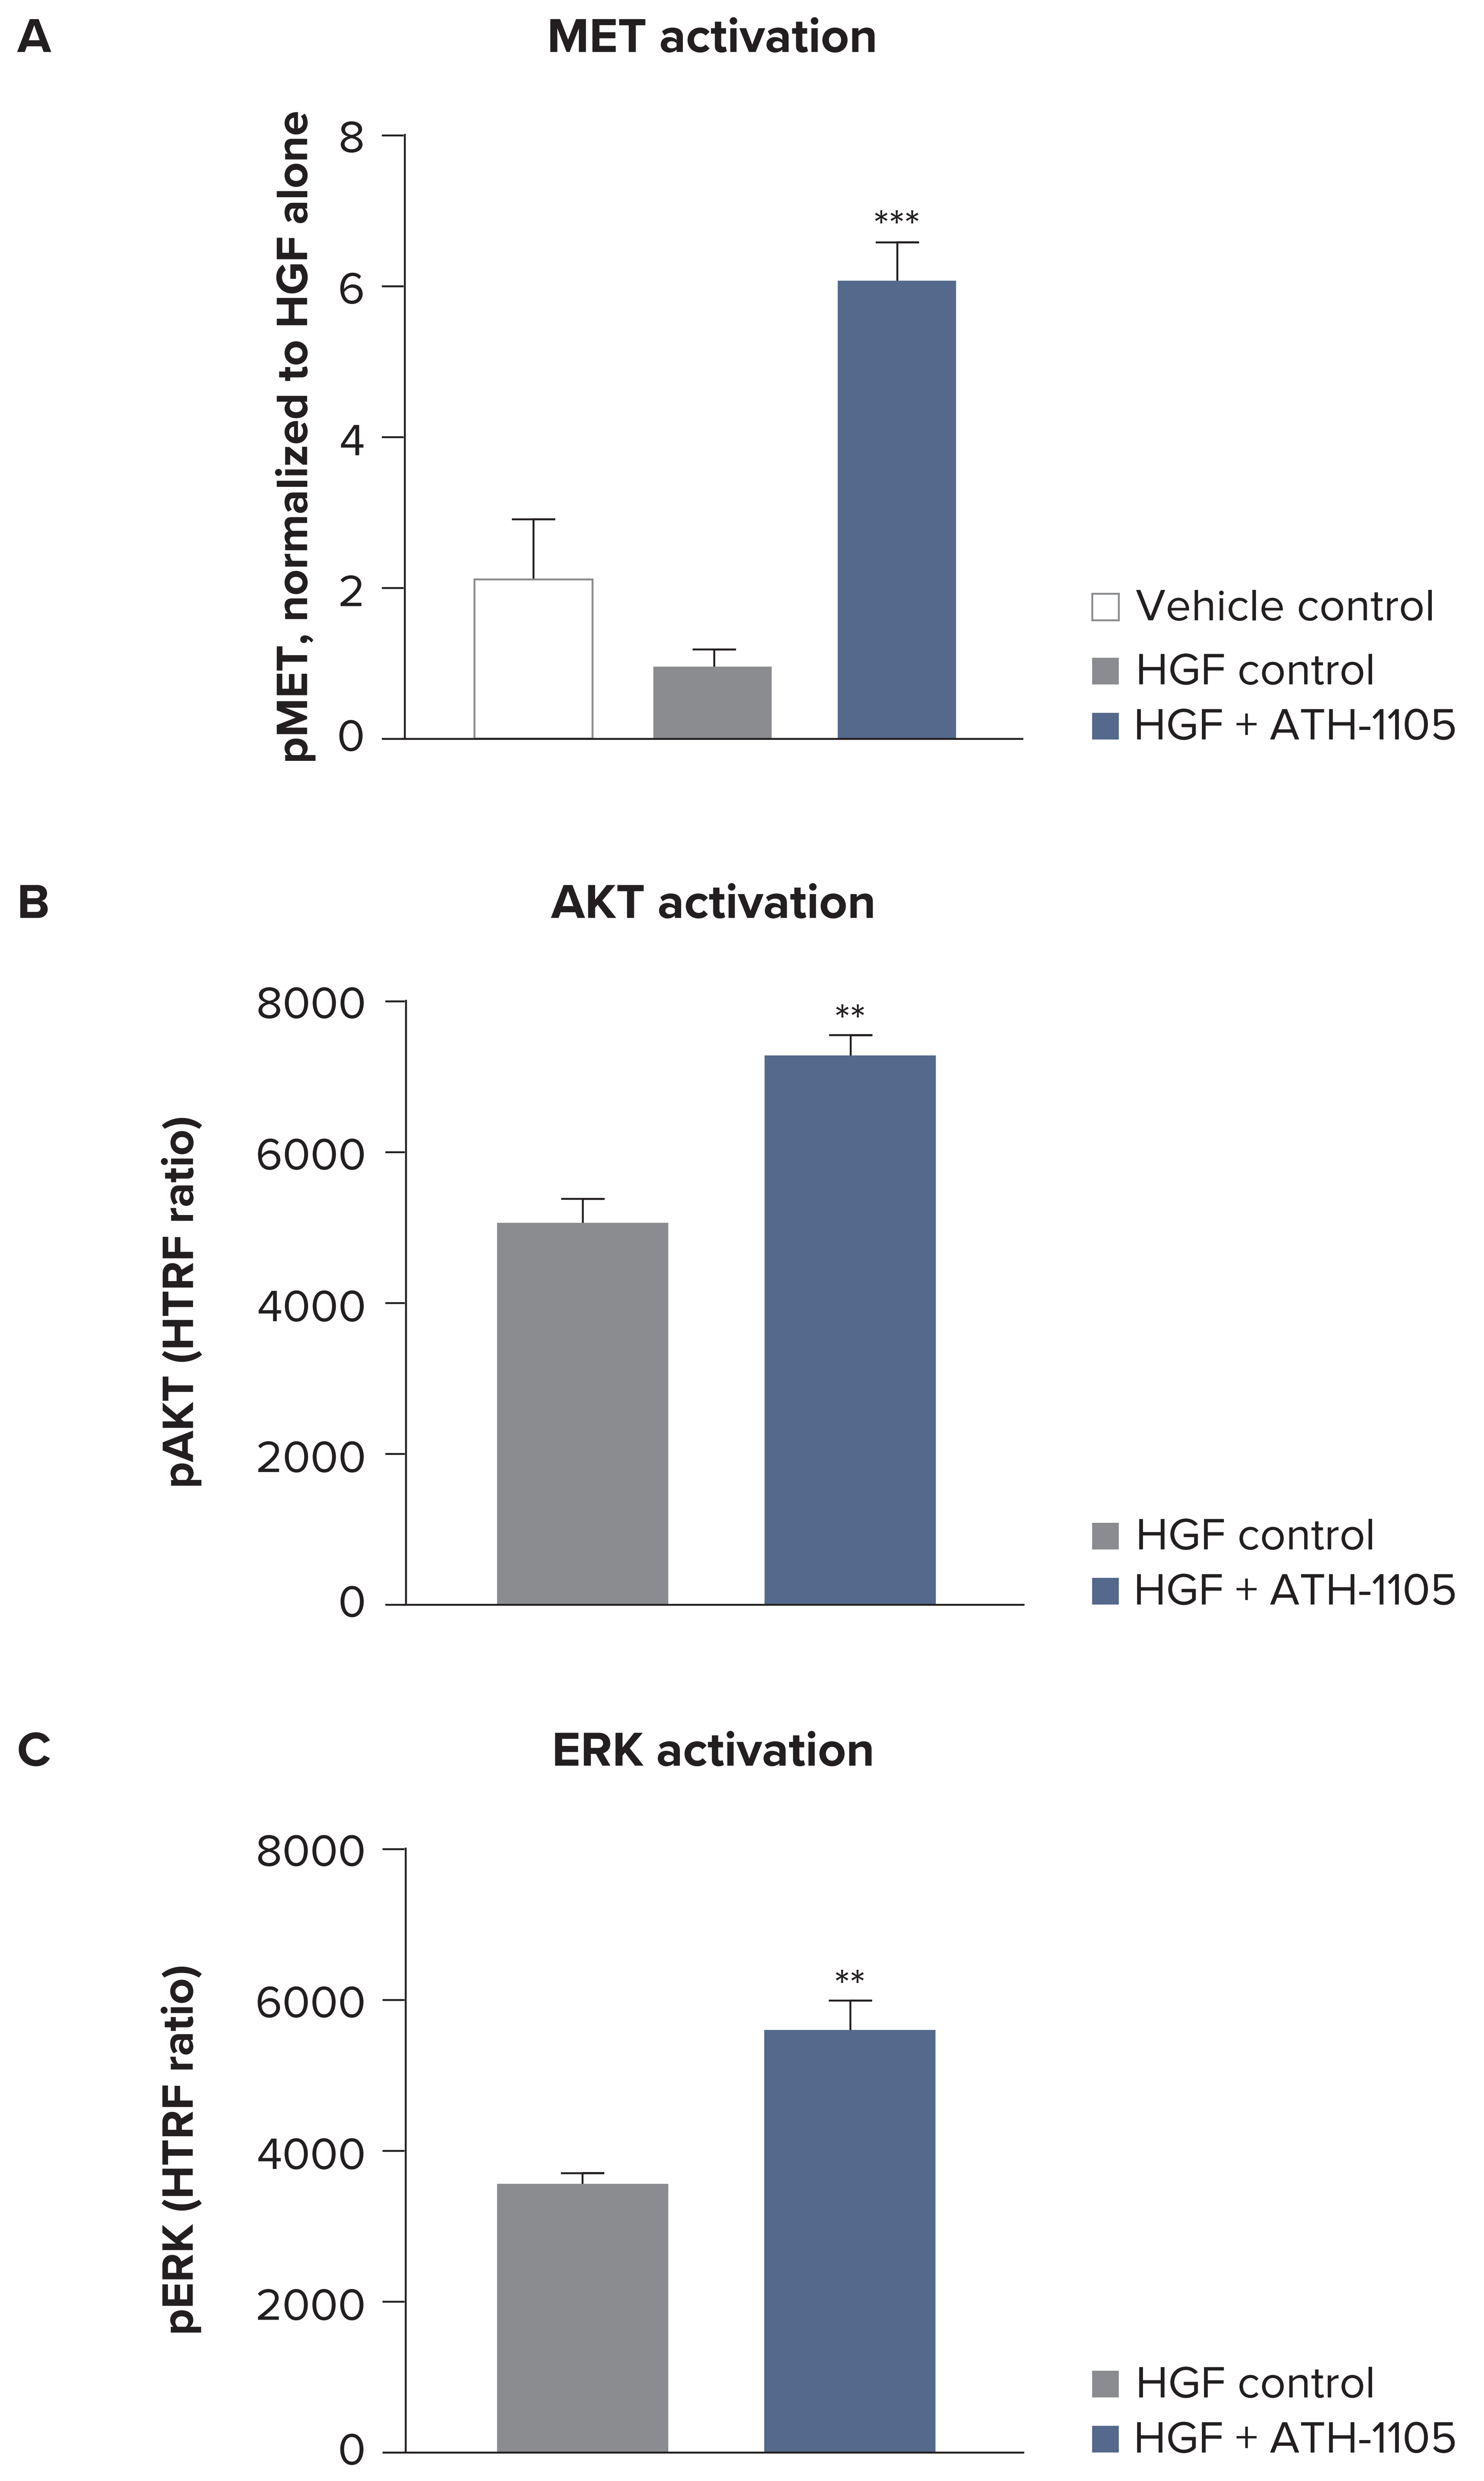
A
MET activation
pMET, normalized to HGF alone
0
2
4
6
8

Vehicle control
HGF control
HGF + ATH-1105
B
AKT activation
pAKT (HTRF ratio)
0
2000
4000
6000
8000
**
HGF control
HGF + ATH-1105
C
ERK activation
pERK (HTRF ratio)
0
2000
4000
6000
8000
**
HGF control
HGF + ATH-1105

Graphical representation of ATH-1105 activation of (A) MET (one-way ANOVA with Dunnett's test vs HGF control), (B) AKT (unpaired *t* test vs HGF control), and (C) ERK (unpaired *t* test vs HGF control).
Data presented as mean + SEM; n = 3 each.
$p < 0.01$; *$p < 0.001$.

Supplemental Figure S2. T-maze paradigm for assessing LPS-induced cognitive impairment in mice

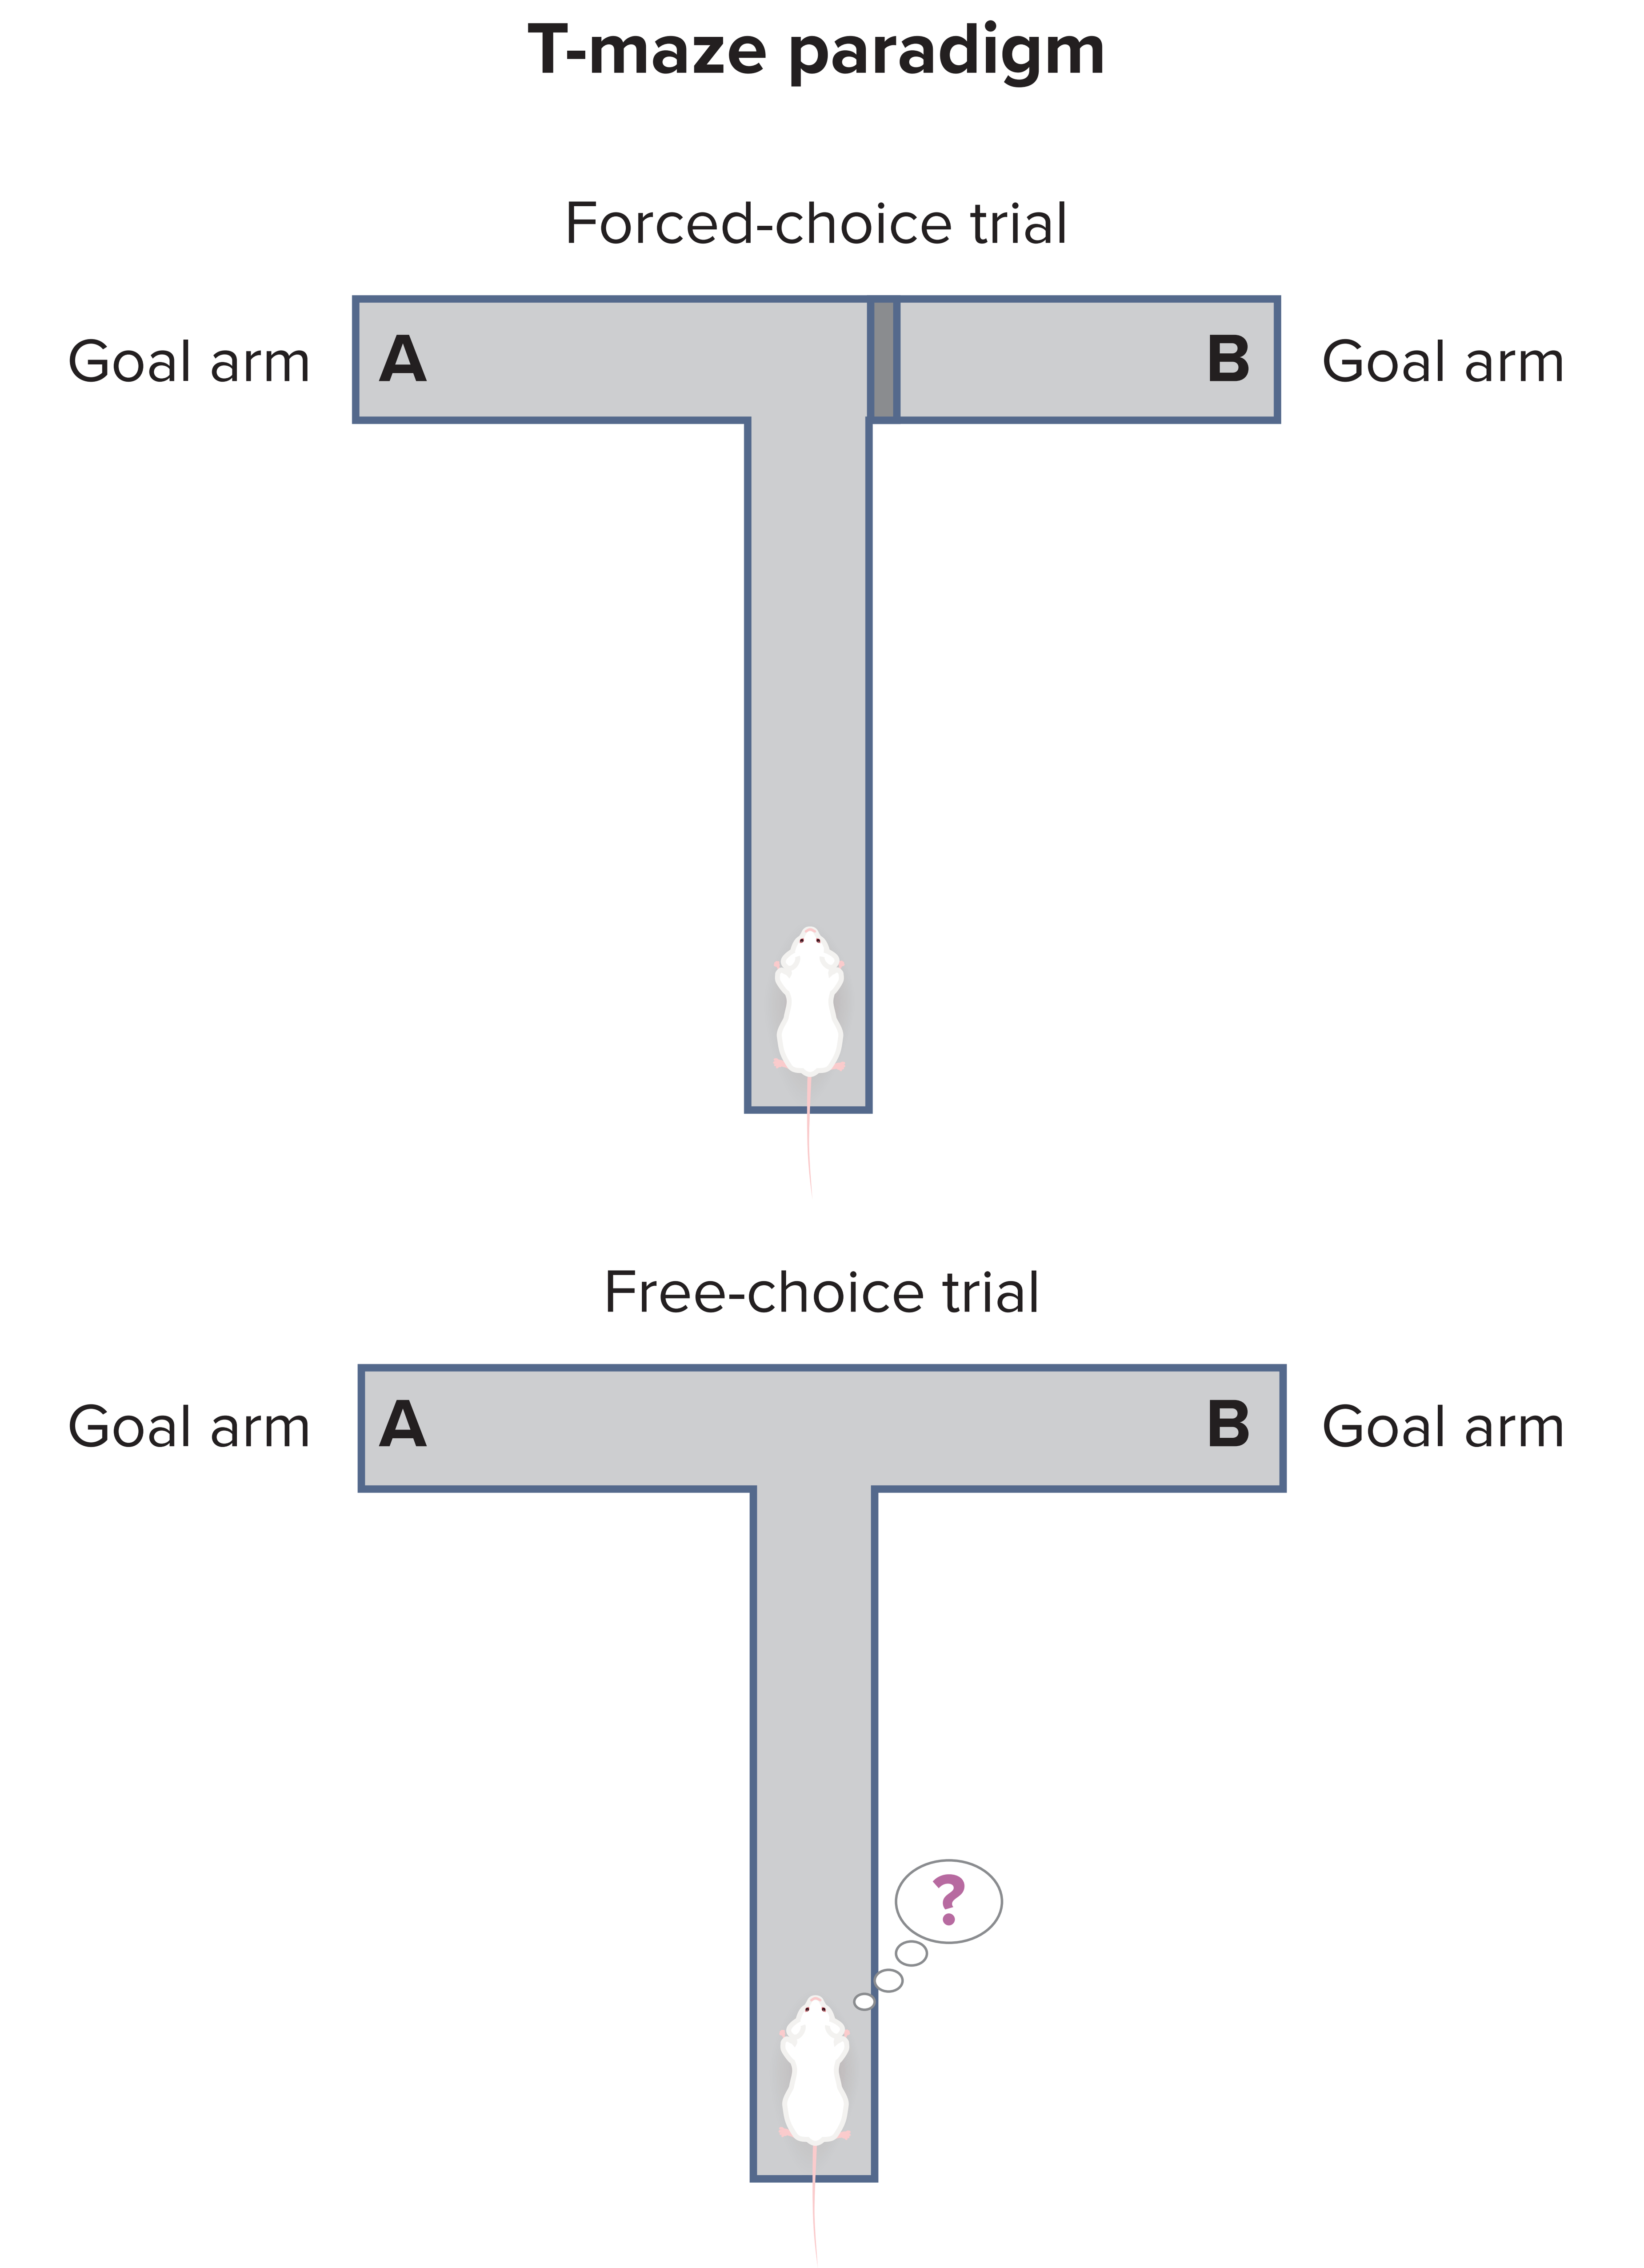
T-maze paradigm
Forced-choice trial
Goal arm
A
B
Goal arm
Free-choice trial
Goal arm
A
B
Goal arm
?

T-maze performance was used to evaluate LPS-induced cognitive impairment. Mice were subjected to one forced-choice trial, in which a door closed off one of the two goal arms, followed by 14 free-choice trials. The rate at which mice chose the alternate goal arm (ie, the one they did not choose during the previous trial) served as an index of working memory performance.

Abbreviations **AKT**, protein kinase B; **ALS**, amyotrophic lateral sclerosis; **ANOVA**, analysis of variance; **ELISA**, enzyme-linked immunosorbent assay; **ERK**, extracellular signal–regulated kinase; **HEK293**, human embryonic kidney 293; **HGF**, hepatocyte growth factor; **HTRF**, homogenous time-resolved fluorescence; **LPS**, lipopolysaccharide; **pAKT**, phosphorylated AKT; **pERK**, phosphorylated ERK; **pMET**, phosphorylated MET; **SEM**, standard error of the mean.

Acknowledgments

This study was sponsored by Athira Pharma, Inc. Medical writing support was provided by ApotheCom and funded by Athira Pharma, Inc.

Disclosures

All authors are employees and stock and/or stock options holders of Athira Pharma, Inc.

Disclaimer

ATH-1105 is an investigational therapy that has not received FDA approval and has not been demonstrated to be safe or effective for any use.

Presented at AAIC 2023; July 16-20, 2023; Amsterdam, Netherlands, and virtual